P038

THE EFFECT OF TIME AND TEMPERATURE DURING TRANSPORTATION OF UMBILICAL CORD TISSUE ON THE VIABILITY, FUNCTIONALITY AND DIFFERENTIATION POTENTIAL OF MESENCHYMAL STEM CELLS FOLLOWING CRYOPRESERVATION AND THAWING

Ann Smith* [1], Francisco Delgado-Rosas[2], Maria Fabra[2], Natalia Aparicio[2], Eva Ainse[2], Alfonso Sanchez[2], Kauser Hussain[1]
[1]Smart Cells International, London, United Kingdom, [2]Ivida, Cord Blood Bank of the IVI Group, Madrid, Spain

European Society for Blood and Marrow Transplantation

smartCells Stem cells for life

ivida

Introduction

There is considerable interest in the potential of mesenchymal stem cells (MSCs) derived from umbilical cord blood (UCB) and more recently cord tissue (CT) for use in regenerative medicine and banking of CT along with UCB. There are recognised procedures to extract and culture viable, functional MSCs pre/post cryopreservation but the effects of transit conditions from collection to laboratory have not been delineated.

Transit conditions are integral to quality assurance to ensure an efficacious end product. This study aimed to define such conditions.

Material and methods

Transit conditions: CT samples (n=15) were transported at approximately 10°C from clinic to laboratory with continual temperature monitoring. In all cases laboratory manipulation of samples commenced 24 hours post cord collection. CT samples, in the original collection medium, were subjected to exposure to constant temperatures of 2°C, 10°C or 28°C for 2, 3, 4 or 5 days. Following decontamination, these CT samples were dissected to yield segments that were either tested fresh or frozen/thawed then tested. Testing comprised viability assessment, phenotypic characterisation and MSC differentiation assays.

Initial cell culture: CT segments (fresh or post freeze/thaw) were cultured for 14 days and following second passage, viable cells typically formed 80-90% confluence and were harvested by trypsinization.

Flow cytometric analysis of harvested cells: positive cell surface markers specific for human MSCs (CD90, CD105 and CD73) and negative (CD11b, CD19, CD34-, CD45 and HLA-DR) were analysed. In all cases MSC phenotype was confirmed.

MSC differentiation assay: In vitro differentiation potential of the harvested MSCs along adipogenic, osteogenic and chondrogenic lineages was undertaken using commercially available induction media StemPro^{R} Osteogenesis, adipogenesis and chondrogenesis kits.

Results

A) FRESH CORD TISSUE SAMPLES

NO = non-viable

Sample	2°C					% viability	10°C					% viability	28ºC					% viability
	1	2	3	4	5		6	7	8	9	10		11	12	13	14	15	
Day 1	First 24 hrs @ 10°C					100	First 24 hrs @ 10°C					100	First 24 hrs @ 10°C					100
Day 2	OK	NO	OK	OK	OK	80	OK	OK	OK	OK	OK	100	OK	OK	Contam	OK	OK	80
Day 3	OK	OK	OK	OK	OK	100	OK	OK	OK	OK	OK	100	OK	OK	OK	OK	NO	80
Day 4	OK	OK	OK	NO	OK	80	OK	NO	NO	OK	OK	60	NO	NO	OK	OK	NO	40
Day 5	OK	OK	NO	NO	OK	60	OK	OK	OK	OK	OK	100	NO	NO	NO	OK	NO	20

B) THAWED CORD TISSUE SAMPLES

NO = non-viable

Sample	2°C					% viability	10°C					% viability	28°C					% viability
	1	2	3	4	5		6	7	8	9	10		11	12	13	14	15	
Day 1	First 24 hrs @ 10°C					100	First 24 hrs @ 10°C					100	First 24 hrs @ 10°C					100
Day 2	OK	OK	OK	OK	OK	100	OK	OK	OK	OK	OK	100	OK	OK	OK	OK	OK	100
Day 3	OK	OK	OK	OK	OK	100	OK	OK	OK	OK	OK	100	OK	OK	OK	NO	OK	80
Day 4	OK	OK	OK	OK	OK	100	OK	OK	Contam	OK	OK	80	OK	NO	NO	NO	OK	40
Day 5	OK	OK	OK	NO	OK	80	OK	OK	OK	OK	OK	100	NO	NO	NO	NO	NO	0

A)

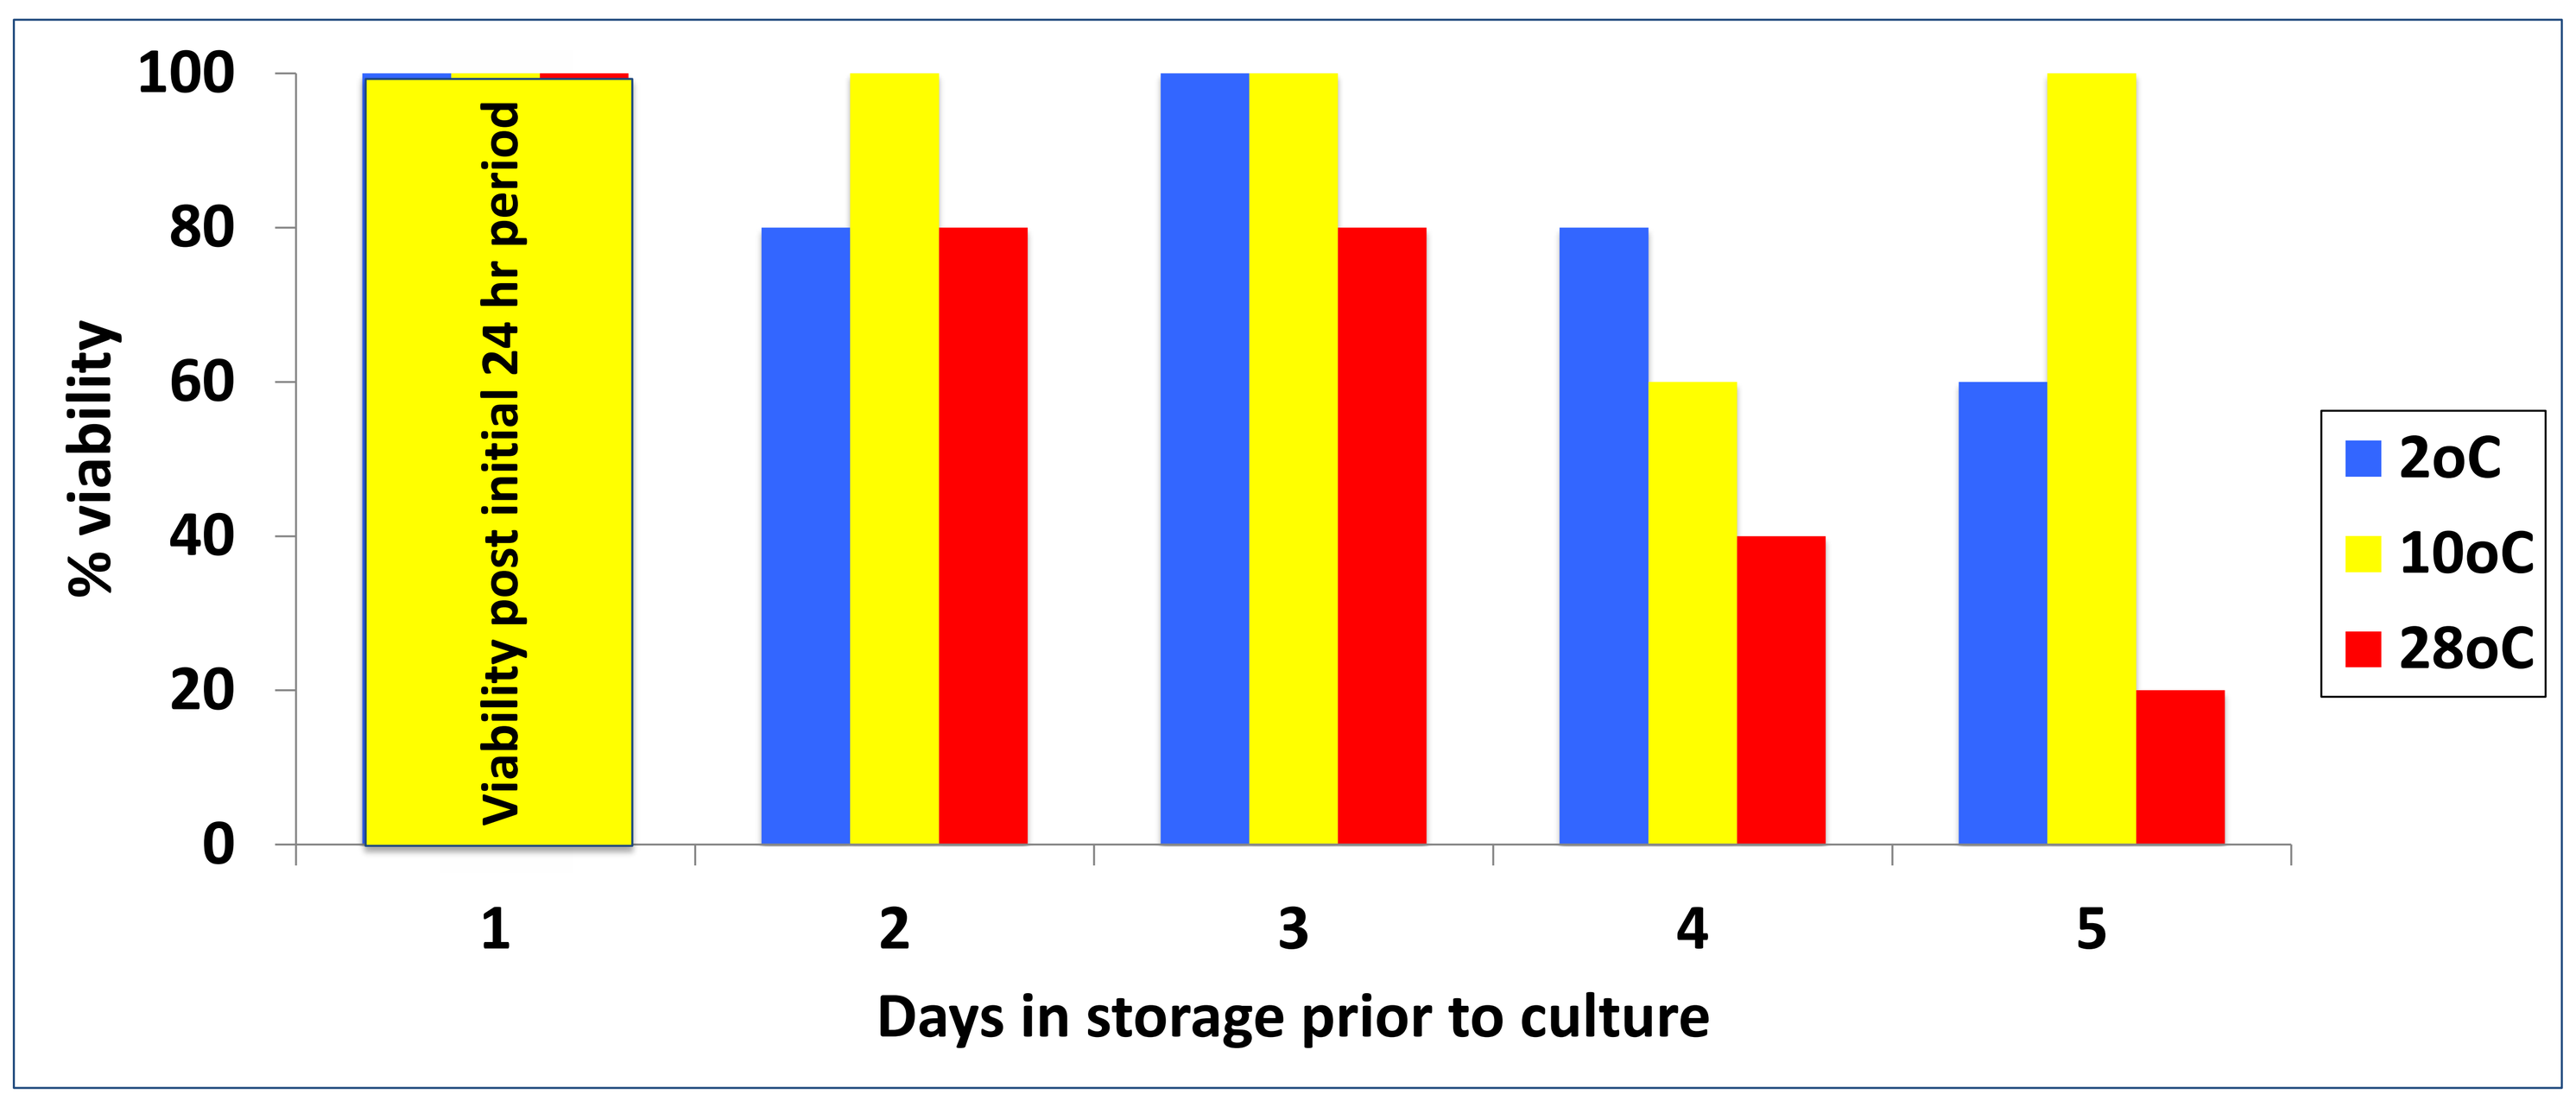
100
80
60
40
20
0
% viability
Viability post initial 24 hr period
1
2
3
4
5
Days in storage prior to culture
2oC
10oC
28oC

B)

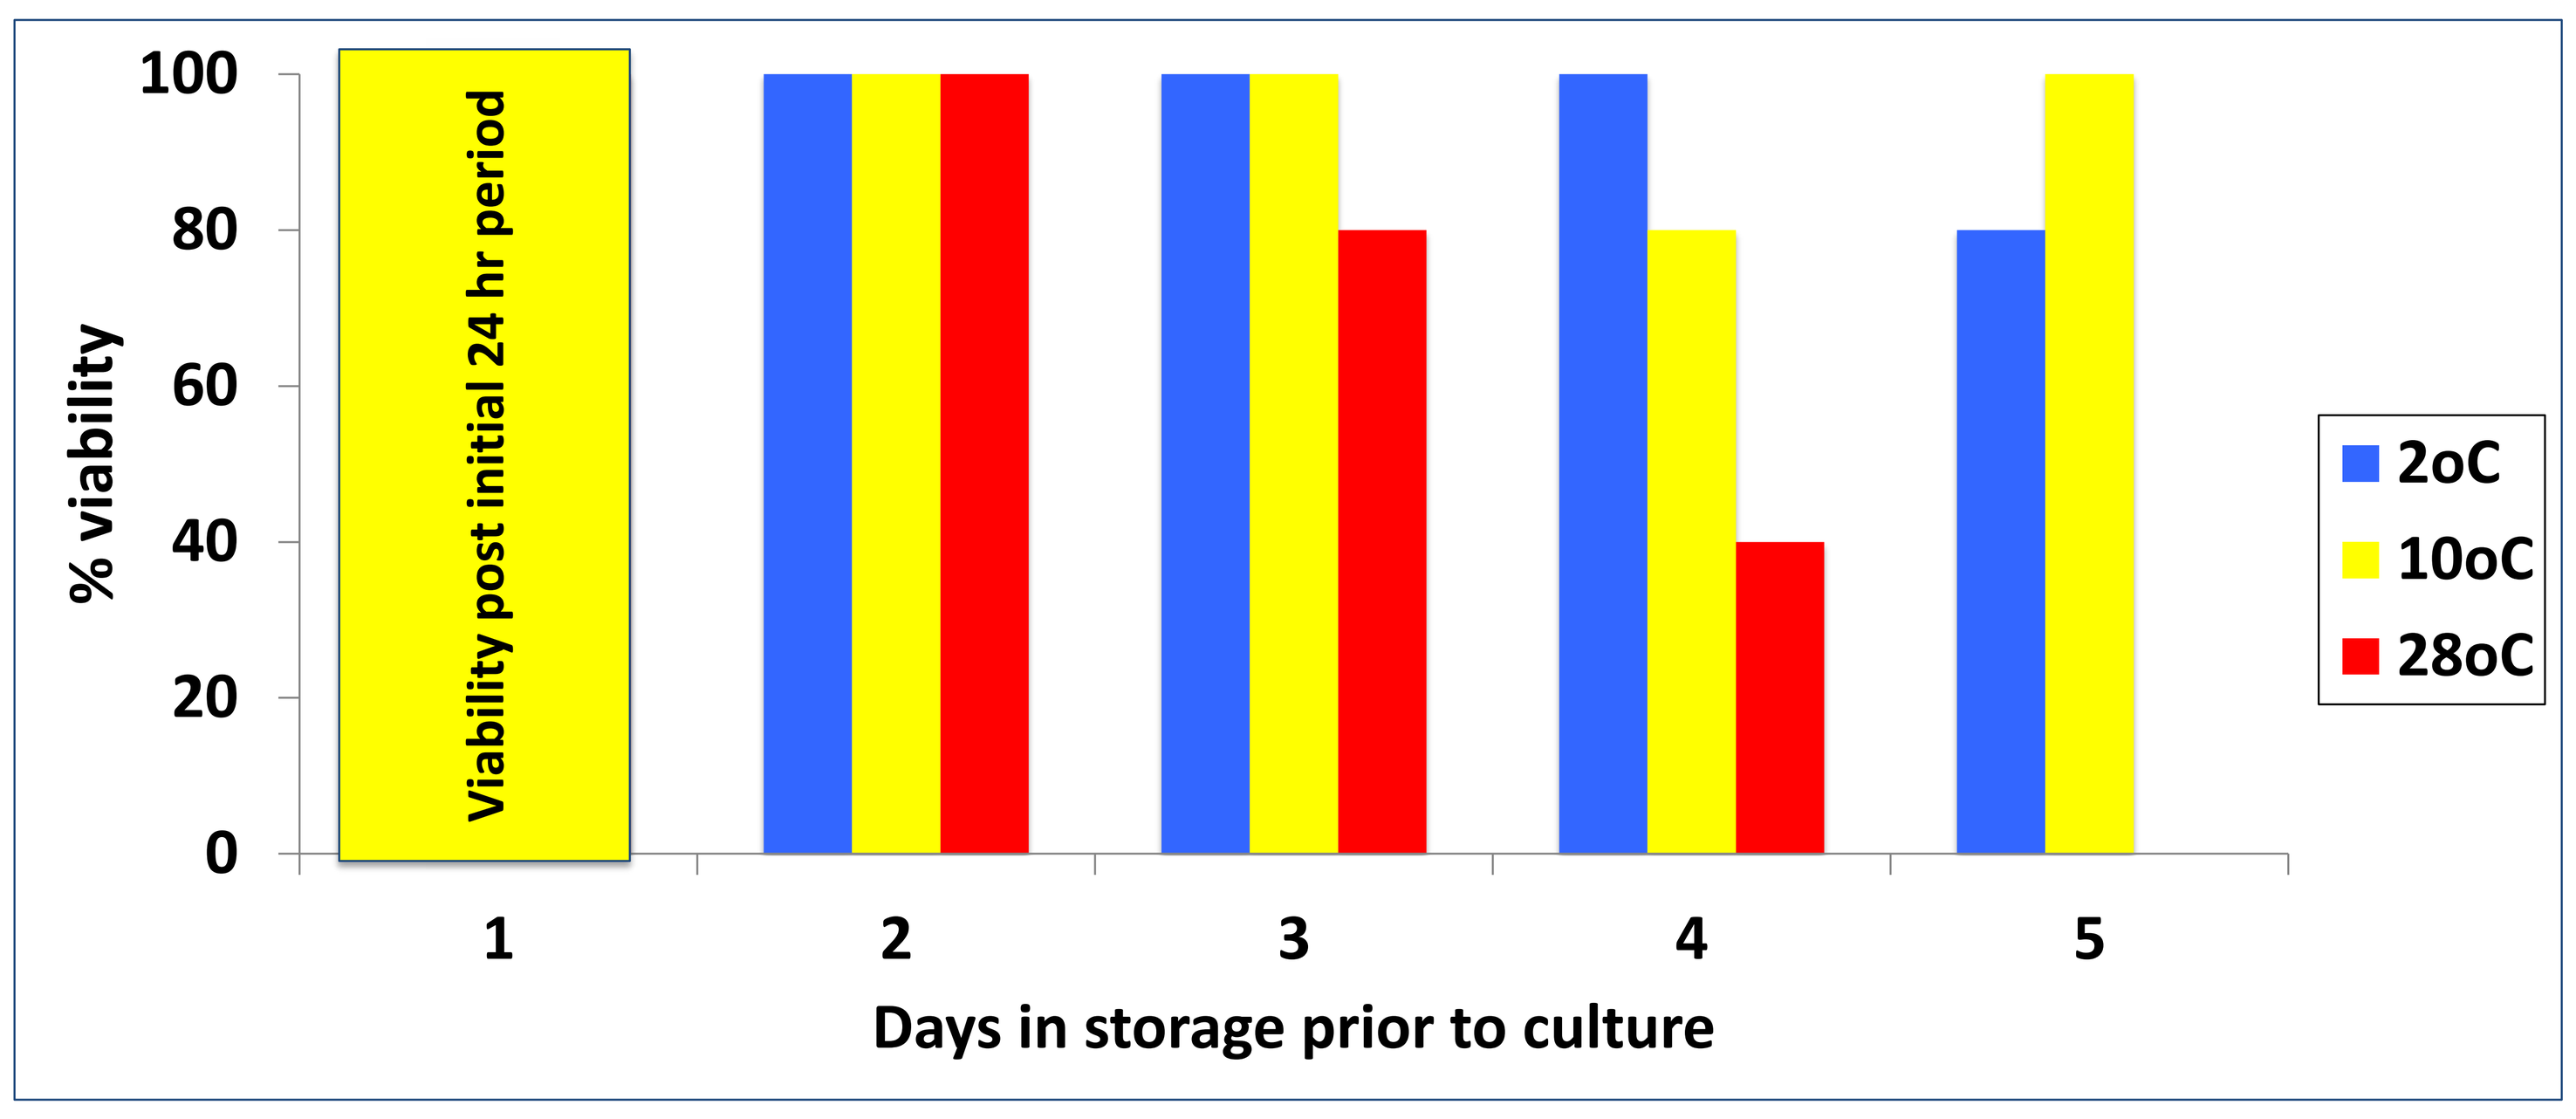
100
80
60
40
20
0
% viability
Viability post initial 24 hr period
1
2
3
4
5
Days in storage prior to culture
2oC
10oC
28oC

Figure 1. Fresh-Thawed CT viability assessment after different days in transit and at different environment conditions. **A)** Overall, viability of MSC precursors in fresh CT at 28°C for all transit times, was significantly impaired ($p \leq 0.05$) compared to no significant adverse effect at 2°C and 10°C ($p = 0.38$ and $p = 0.069$ respectively). At 28°C, viability levels were 80%, 40% and 20% after 3, 4 and 5 days respectively. **B**) Following thawing and subsequent culture, CT that had been stored at 2°C for 4 and 5 days exhibited viability of 100% and 80% respectively. At 10°C, 100% viability was apparent for all time points. Overall, storage at 28°C significantly affected the ability to generate viable cells from cryopreserved CT, resulting in viability levels of 80%, 40% and 0% for 3, 4 and 5 days ($p \leq 0,01$).

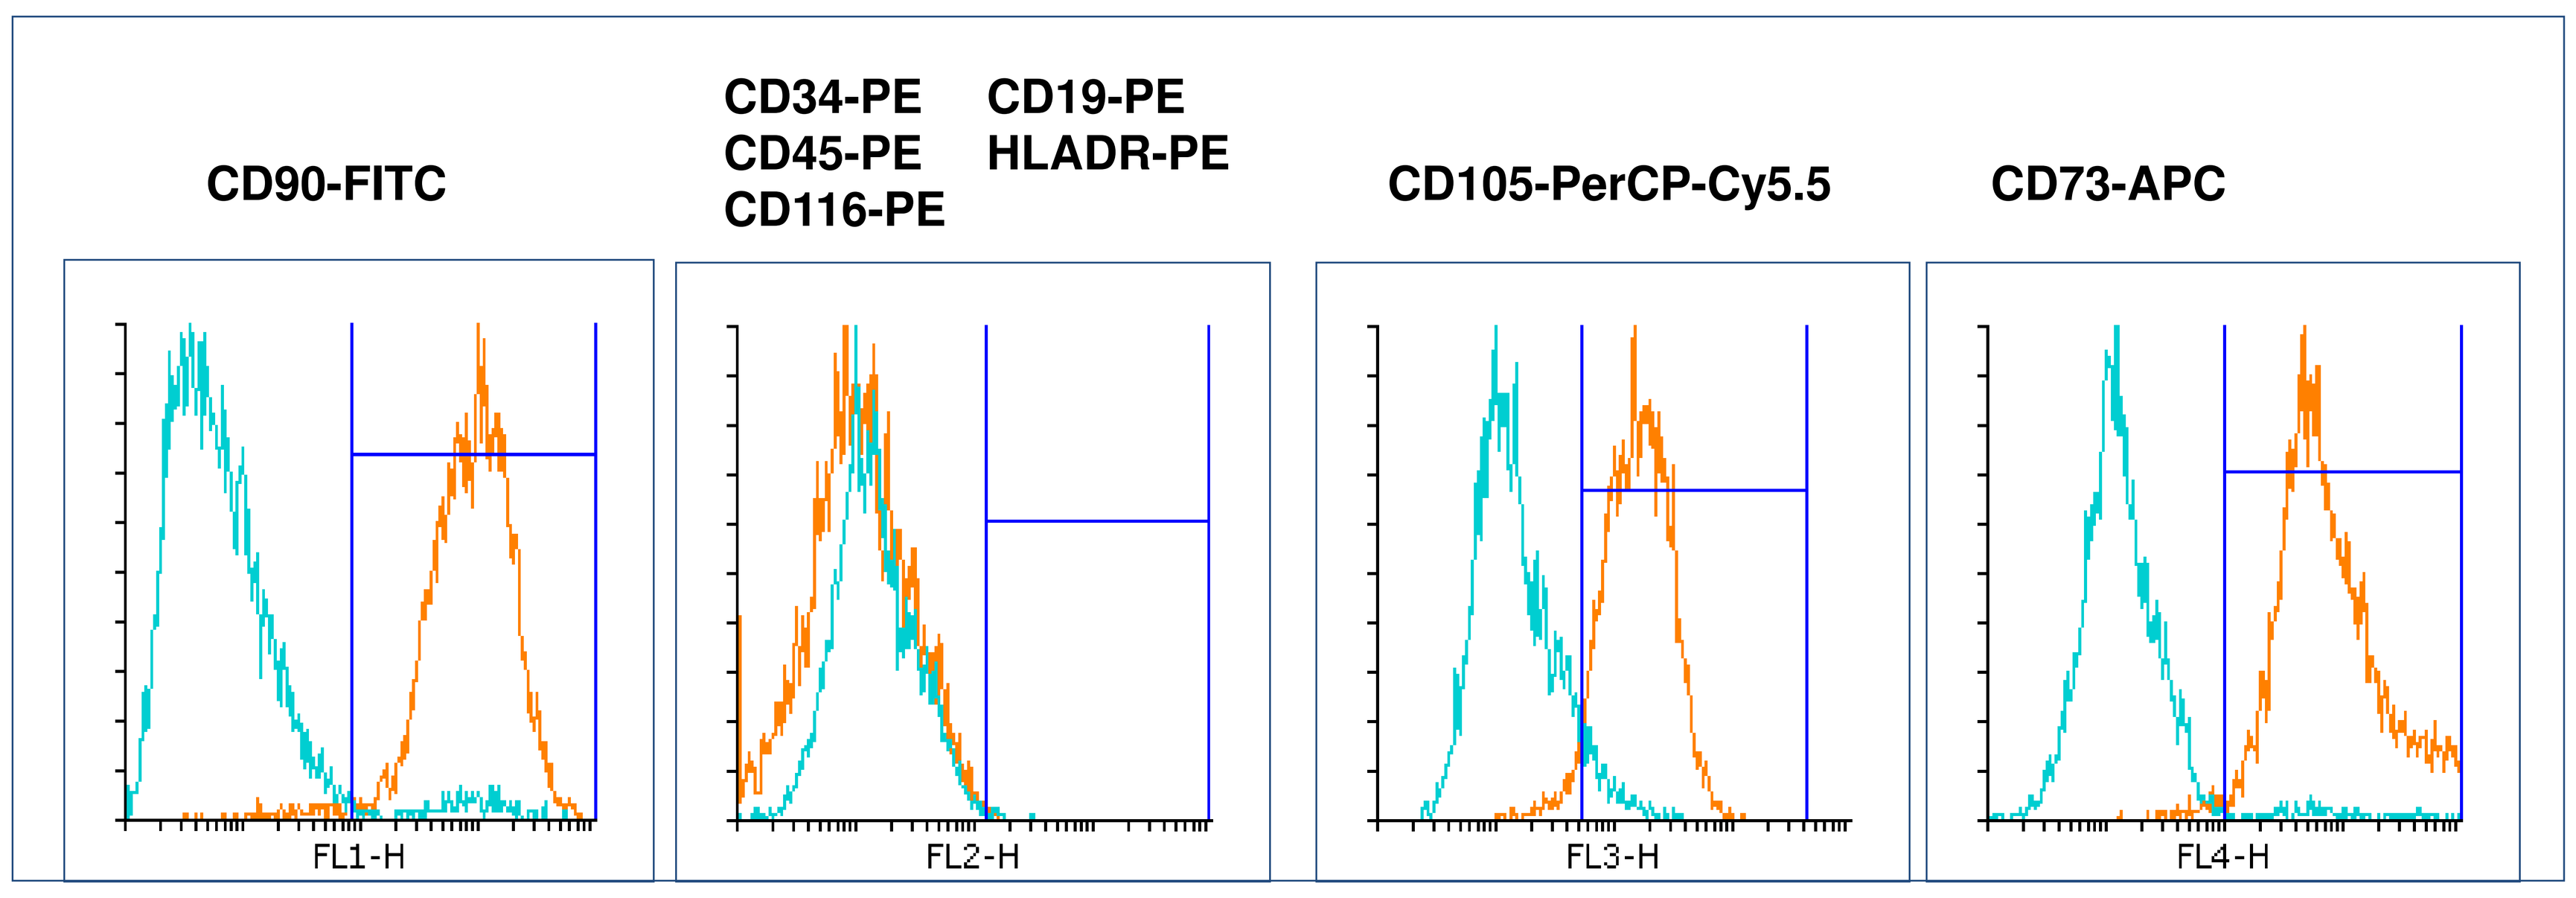
CD90-FITC
CD34-PE
CD45-PE
CD116-PE
CD19-PE
HLADR-PE
CD105-PerCP-Cy5.5
CD73-APC
FL1-H
FL2-H
FL3-H
FL4-H

FL1-FITC	%Gated		FL2-PE	%Gated		FL3-PerCP	%Gated		FL4-APC	%Gated
Isotype	3,87		Isotype	0,38		Isotype	7,55		Isotype	3,65
+ Marker	97,47		- Marker	0,52		+ Marker	95,08		+ Marker	98,04

Figure 2. Typical flow cytometric plots and associated data for MSC cell surface markers from cells derived from CT following storage for 4 Days at 28°C. Plots show expression histograms for isotype controls (blue) and antibody staining corresponding to the markers analysed (orange). Positive expression regions were established from isotype controls. Data is tabulated as % positivity for each marker combination. In all cases,

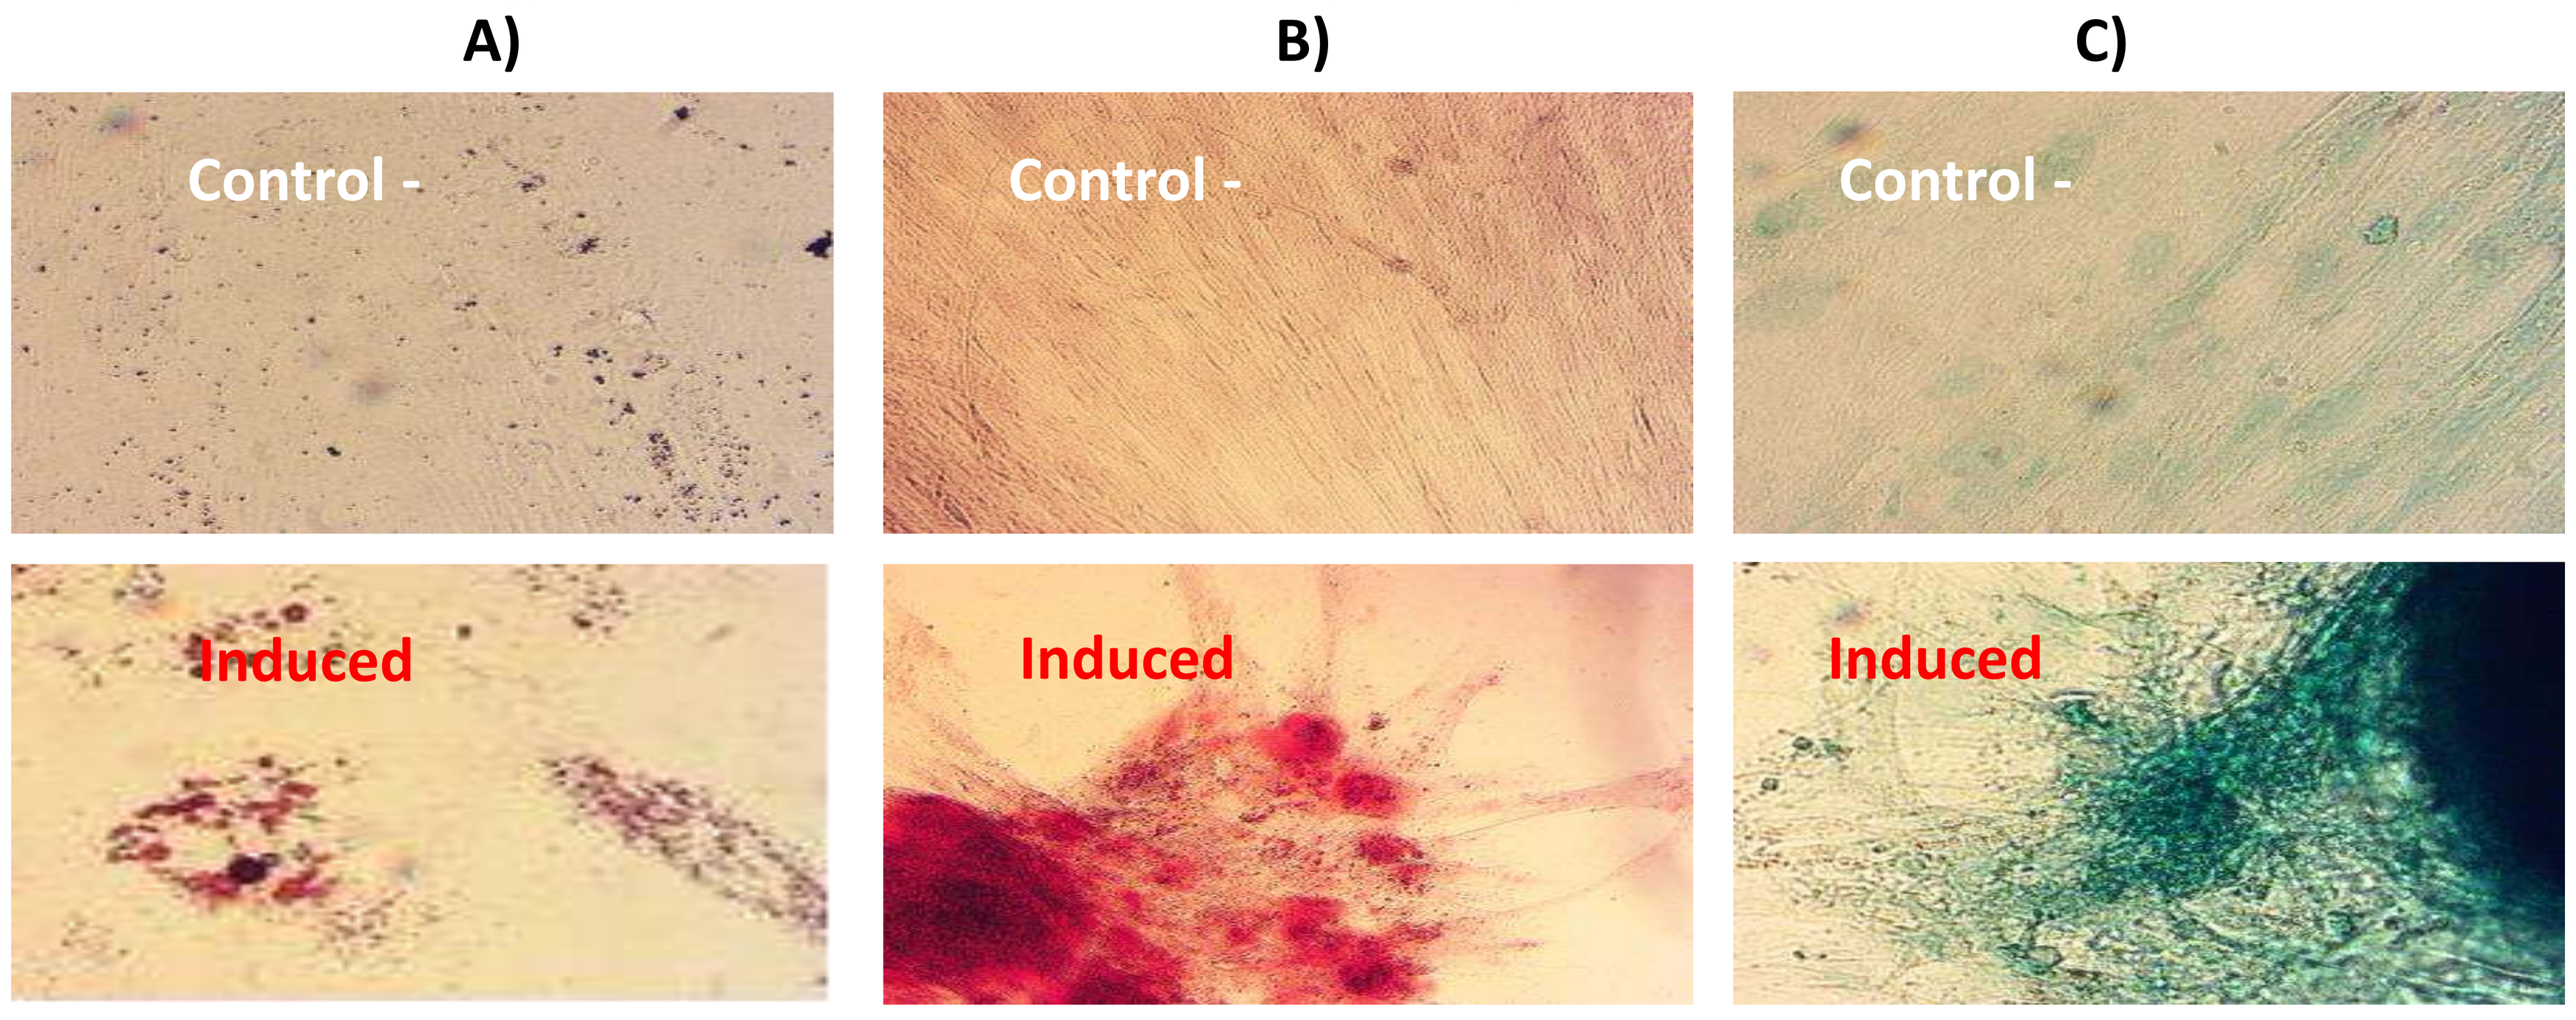
A)
B)
C)
Control -
Control -
Control -
Induced
Induced
Induced

Figure 3. Colourimetric testing for the induction of differentiation of mesenchymal cells into adipocytes, osteocytes and chondrocytes. The figure relates to MSCs derived from CT that had been stored for 4 Days at 28°C . The controls for each assay were mesenchymal cells to which no specific induction culture medium was added (CONTROL -). Controls did not show positive staining and cultures maintained fibroblast morphology whereas the induced cultures exhibited specific morphologies and stained appropriately. A) ADIPOGENESIS: induced cells contained lipid droplets stained with Oil Red. B) OSTEOGENESIS: differentiated cells exhibited positive Alizarin Red S staining of calcium deposits. C) CHONDROGENESIS: induced cells displayed blue staining caused by the dye Alcian Blue in the extracellular matrix, due to the formation of cartilage compounds.

Conclusions

This study demonstrates no significant adverse effect on the generation of viable and functional MSCs from frozen/thawed CT samples following up to 5 days with initial storage/transit while fresh at 2°C or 10°C with 10°C being optimal. Cells following fresh storage/transit at 28°C for 3 days recovered after freeze/thawing with 80% viability and were phenotypically typical of mesenchymal cells exhibiting adipocytic, osteocytic and chondrocytic differentiation potential. JACIE V6 standards state that the ideal transport temperature may range from 2-24°C and that most products should not be transported at temperatures above 24°C. Based on the data presented, and taking into account the JACIE recommendation, it would therefore seem reasonable to aim to transport CT at temperatures not greater than 24°C (as this is safely below 28°C) and transit times not in excess of 3 days.

CT derived MSCs are being increasingly considered for indications in the growing field of regenerative medicine and also for their ability to modulate the immune response. This study has delineated permissible time lapse from collection to storage of CT and the range of transit temperatures over which samples are able to yield viable MSCs that retain their biological and regenerative properties.

#EBMT16

http://www.smartcells.com/

http://www.bancodecordonivida.com/

www.ebmt.org